Impact of single nucleotide polymorphisms in human AOX1 on the enzymatic activity of aldehyde oxidase in cryopreserved human hepatocytes

Scott Heyward[1], J. Matthew Hutzler[2], Young-Sun Yang[3], Timothy Moeller[1], Hadassa Guttman[1], Caitlin Brown[1]

[1]BioreclamationIVT, Baltimore, MD, [2]Quintiles, Indianapolis, IN, [3]Boehringer-Ingelheim Pharmaceuticals Inc.; Medicinal Chemistry, Drug Discovery Support (DMPK) Ridgefield, CT .

Abstract

Aldehyde oxidase (AO) is a molybdenum cofactor containing enzyme localized in the cytosol with relatively high expression levels in human hepatocytes. It is of increasing importance for drug development with an increasing number of new chemical entities (NCE) found to be substrates. Hepatic expression shows a high amount of variation among the major preclinical species and between genders in some of those species. Exploration of systems to properly assess the role of aldehyde oxidase in the stability of NCEs has led to the use of human liver cytosol and intact cryopreserved human hepatocytes. These systems have show large variation in enzyme expression and catalytic capacity. Exploration of genetic variants in the hAOX1 gene has exposed the potential contribution of a number of variants to this variability. We explored, in cryopreserved human hepatocytes, the prevalence and impact of a group of single nucleotide polymorphisms (SNP) in hAOX1 on AO activity. We generated genotypes for seven SNPs in the hAOX1 gene and analyzed the haplotypes for frequency and racial distribution in a population of 221 individual human liver donors. Incubations with carbazeran were performed with a subset of hepatocyte lots and in vitro intrinsic clearance values were generated. Values were analyzed for influence of age, sex, BMI and race. Genotypes for four of the SNPs were monomorphic in our population. Three SNPs were detected and showed allelic frequencies in our racial populations similar to reported values. These SNPs were analyzed for correlation to clearance of carbazeran. The SNP rs55754655 (3404A>G) was present but no statistical difference in activity was seen between genotypes. Those donors containing rs3731722 (3890A>G) mutant alleles showed a statistically significant increase in carbazeran clearance (224.2 (wildtype) vs 302.2 (containing G) ml/min/kg, P= .02326). However, for the intronic SNP rs10931910, when haplotypes containing rs3731772 were excluded, a significant reduction was seen in activity in the mutant populations, (279.2 (wildtype) vs 212.2 (containing G) ml/min/kg, P= .03789). These results help to explain a portion of the variation seen in cryopreserved human hepatocytes, however exploration of larger donor pools, additional genetic variants, and a deeper investigation of non-genetic factors may be necessary to fully characterize the source of diversity in donors.

Introduction

Aldehyde Oxidase has gained note as an important non-cytochrome 450 oxidative pathway due to the increase in pharmaceuticals in development which are substrates and the unexpected failure of compounds in clinical trials.(Akabane 2011, Dittrich, 2002) This has been in part due to inadequate preclinical animal species testing where significant differences in aldehyde oxidase expression exist. This has let to the development of predictive *in vitro* models for the contribution of aldehyde oxidase to drug metabolism. In this development a high variability has been seen in human liver derived reagents.(Fu, 2013, Hutzler 2014). The source of the variability has not been fully characterized, however, genetic variation may play a part. A small number of studies have explored the impact of genetic polymorphisms in the AOX1 gene on the activity of aldehyde oxidase or in clinical outcomes where aldehyde oxidase may play a metabolic role.(Hartmann, 2012, Ramirez, 2013, and Smith, 2009). In Ramirez et. al.(2013) and Smith et. al.(2009) no conclusive association was seen in studies with clinical populations, possibly due to study size. We have utilized isolated hepatocytes in an *in vitro* system to characterize the impact of genotype. We used carbazeran as a probe substrate which has shown high turnover by aldehyde oxidase *in vitro*. (Hutzler, 2012) In this study we explored the prevalence of seven SNPs in AOX1 in our liver donor populations. The presence of these SNPs was assessed for impact on carbazeran *in vitro* clearance. We hope that our analysis of the contribution of donor demographics and genetic background will better explain the variability of aldehyde oxidase activity in human hepatocytes and provide a more predictive model for drug clearance in the clinical setting.

Methods

Cryopreserved Human Hepatocytes:

Hepatocyte Incubations: Cryopreserved human hepatocytes were obtained from BioreclamationIVT and thawed according to manufactures protocols. Briefly, vials were warmed in a 37C water bath until just thawed and transferred to 45mls of prewarmed InVitroGro HT media, spun at 50xg for 5min and resuspended in KHB buffer. Cells were assessed for viability and number by the trypan blue exclusion method. Cells were diluted to 1x10^6cells/ml(a 2x solution) and 250,000 cells transferred to a 48-well plate which was placed at 37C. Reactions were started by the addition of substrate at a final concentration of 1uM and incubated at 37C on a shaker at 200RPM. At timepoints of 0, 5, 15, 30, 60, 120min 50ul of sample was transferred to stop solution consisting of 150ul of acetonitrile (1% acetic acid) with .25uM tolbuamide as an internal standard. Analysis was performed by LC MS/MS

Single Nucleotide Polymorphism Analysis.

Genomic DNA was prepared from liver donor samples, either tissue or cryopreserved hepatocytes (n = 221 donors) using the Qiagen QIAamp DNA Mini kit (Gaithersburg, MD). TaqMan Genotyping SNP assays and TaqMan Genotyping Master Mix were obtained from Life Technologies (Carlsbad, CA) and were used in reactions according to the manufacturer's instructions. Reactions were run on an ABI7500 Fast Real-Time polymerase chain reaction system, and data were captured and analyzed using SDS v1.3 software. All SNP markers detected were in Hardy-Weinberg equilibrium as tested by Fisher's exact test. Statistical analysis was performed using GraphPad Prism (v5.02) (La Jolla, CA).

Intrinsic Clearance Estimates.

In vitro intrinsic clearance (Clint) was estimated (ml/min per kilogram) after incubations in human hepatocytes for 2 hours using substrate depletion methods and the following equation:

Clint =.693 X 1 / t $_{1/2}$ X ml incubation /hepatocytes X gm liver wt /kg body wt X hepatocytes/gm liver wt

where t1/2 is the observed in vitro substrate depletion half-life (min) as determined by linear regression. Scaling factors for intrinsic clearance calculations were as follows: 25.7 g liver weight per kilogram of body weight and 120 x10^6 hepatocytes per gram of liver.

Statistical Analysis

Genetic polymorphisms and carbazeran clearance values were associated for the individual donors and analyzed using GraphPad Prism (v5.02)(La Jolla CA). Statistical analysis was performed for allelic variation using chi square analysis and t-test for correlation between activity and genotype. Whisker plots were generated with mean+/-SEM displayed.

References:

Akabane T, Tanaka K, Irie M, Terashita S, and Temaura T (2011) Case report of extensive metabolism by aldehyde oxidase in humans: Pharmacokinetics and metabolite profile of FK3453 in rats, dogs , and humans. *Xenobiotica*, 41(5); 372-384.

Dittrich Ch, Greim G, Borner M, Weigang-Kohler K, Huisman H, Amelsberg A, Ehret A, Wanders J, Hanauske A, and Fumoleau P.(2002) Phase I and pharmacokinetic study of BIBX1382 BS, an epidermal growth factor receptor (EGFR) inhibitor, given in a continuous daily oral administration. *Eur J Cancer*, 38:1072-1080.

Fu C, Di L, Han X, Soderstrom C, Snyder M, Troutman MD, Obach RS, Zhang H. (2013)Aldehyde oxidase 1 (AOX1) in human liver cytosols: quantitative characterization of AOX1 expression level and activity relationship. *Drug Metab Dispos*. 2013 Oct;41(10):1797-804

Hartmann, T., Terao, M., Garattini, E., Teutloff, C., Alfaro, J. F., Jones, J. P., & Leimkuhler, S. (2012). The Impact of Single Nucleotide Polymorphisms on Human Aldehyde Oxidase. *Drug Metabolism and Disposition*, *40*(5), 856–864.

Hutzler JM1, Yang YS, Albaugh D, Fullenwider CL, Schmenk J, Fisher MB. (2012) Characterization of aldehyde oxidase enzyme activity in cryopreserved human hepatocytes. *Drug Metab Dispos*. 2012 Feb;40 (2):267-75.

Hutzler JM1, Yang YS, Brown C, Heyward S, Moeller T. (2014) Aldehyde oxidase activity in donor-matched fresh and cryopreserved human hepatocytes and assessment of variability in 75 donors. .*Drug Metab Dispos*. Jun;42 (6):1090-7

Ramírez J, Kim TW, Liu W, Myers JL, Mirkov S, Owzar K, Watson D, Mulkey F, Gamazon ER, Stock W, Undevia S, Innocenti F, Ratain MJ.(2014) A pharmacogenetic study of aldehyde oxidase I in patients treated with XK469. *Pharmacogenet Genomics*. Feb;24(2):129-32

Smith, M. A., Marinaki, A. M., Arenas, M., Shobowale-Bakre, M., Lewis, C. M., Ansari, A., … Sanderson, J. D. (2009). Novel pharmacogenetic markers for treatment outcome in azathioprine-treated inflammatory bowel disease. *Alimentary Pharmacology & Therapeutics*, *30*(4), 375–384.

Tayama Y, Miyake K, Sugihara K, Kitamura S, Kobayashi M, Morita S, Ohta S and Kihira K., (2007) Developmental changes of aldehyde oxidase activity in young Japanese children.*Clin Pharmacol Ther*. 81(4):567-72.

Results

Figure 1.

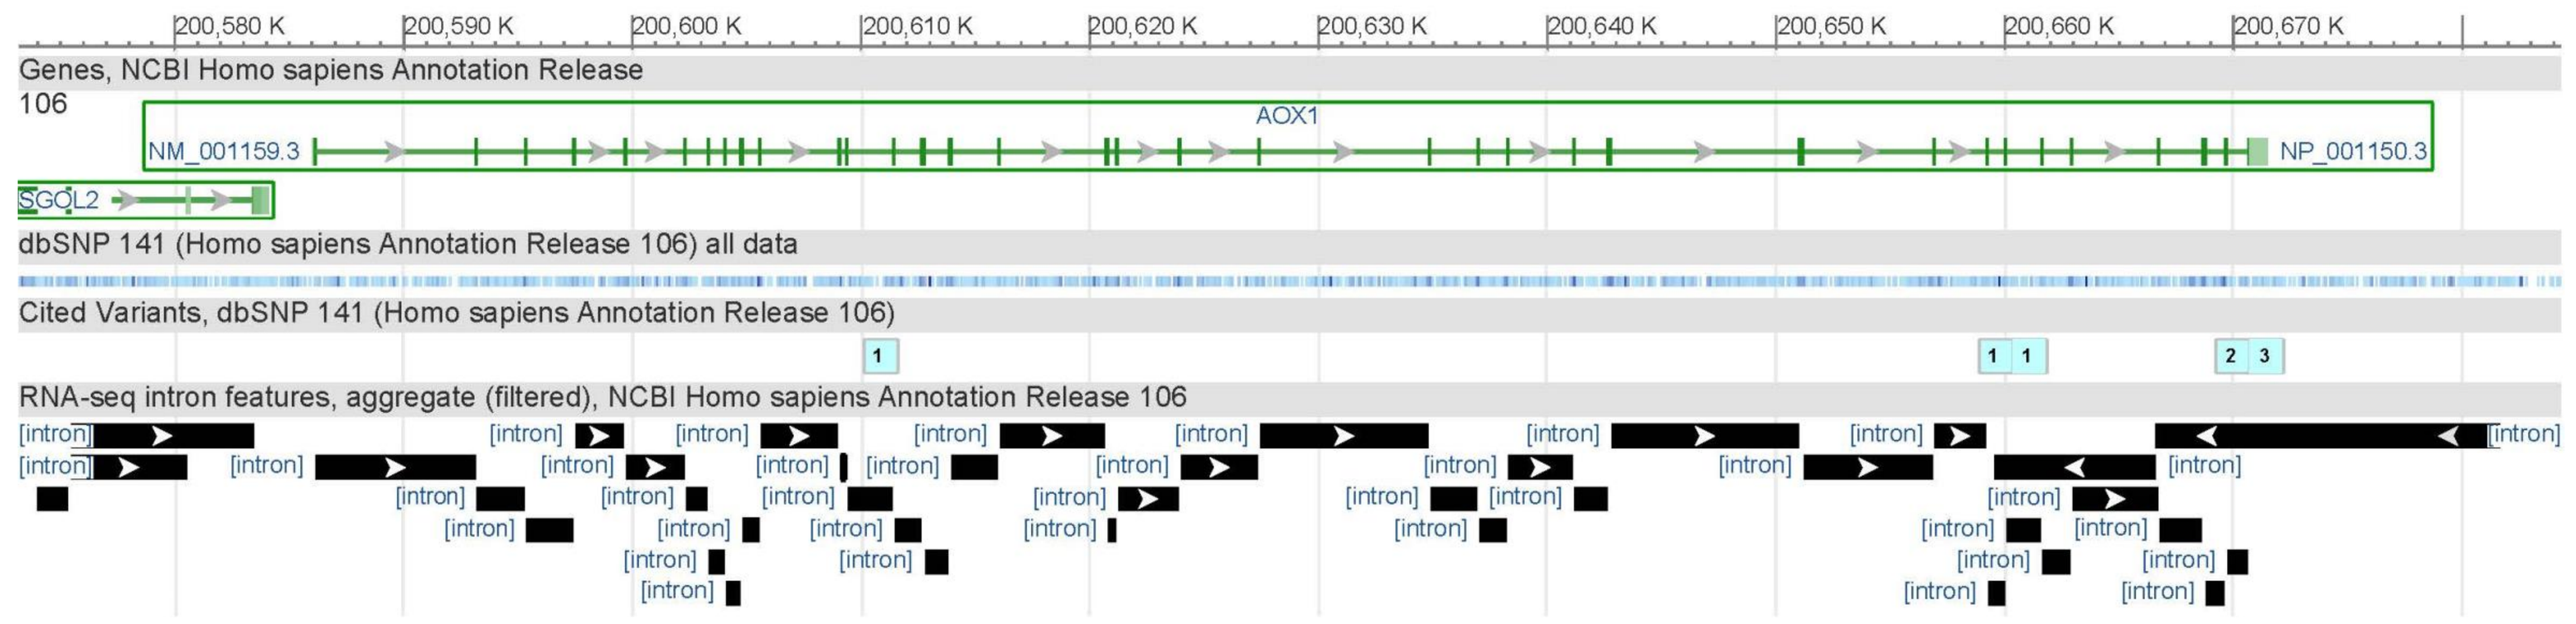
Genes, NCBI Homo sapiens Annotation Release 106
AOX1
NM_001159.3
NP_001150.3
SGOL2
dbSNP 141 (Homo sapiens Annotation Release 106) all data
Cited Variants, dbSNP 141 (Homo sapiens Annotation Release 106)
RNA-seq intron features, aggregate (filtered), NCBI Homo sapiens Annotation Release 106

Annotated view of the human AOX1 gene (Aldehyde Oxidase 1) from GRCh38 Primary assembly describing nucleotide position and gene structure. AOX1 is located on chromosome 2 and contains 35 exons. Exons are indicated by vertical green lines, introns as black blocks below. (NCBI [Internet]. Bethesda (MD): National Library of Medicine (US). [1946] - [cited 2014 Sept 31]. Available from: http://www.ncbi.nlm.nih.gov/)

Table 1.

Reference SNP	Exon	Nucleotide Change	Amino Acid Change	Cohort Minor Allelic Frequency	dbSNP Minor Allelic Frequency
rs10931910	Intron 27	A>G	N/A	.466 [1]	.475 [3]
rs55754655	30	3404A>G	N1135S	.153 [1]	.189 [3]
rs3731722	34	3890A>G	H1297R	.075 [1]	.056 [3]
rs199984835	5	378C>G	Y126Stop	0 [1]	.026 [4]
rs41309768	22	2404C>T	R802C	0 [2]	.006 [4]
rs56199635	25	2762G>A	R921H	0 [2]	.003 [4]
rs141786030	34	3812C>T	S1271L	0 [2]	0 [3]

[1] n=380, [2] n=254, [3] ESP_Cohort_Population n=4552, [4] COLL2006 n=360, n=chromosomes samples analyzed

Summary of single nucleotide polymorphisms (SNP) and allelic frequencies observed and reported. Liver donors were genotyped for the above SNPs using Taqman SNP Assays and allelic frequencies generated. Data is compared to those reported in the NCBI database. Frequencies were in agreement with those reported, with the exception of rs199984835, rs41309768, rs56199635 which were monomorphic in our study.

Figure 2.

		rs10931910		rs55754655		rs3731722	
Race	Genotype	n=	Frequency	n=	Frequency	n=	Frequency
AA	G/G	5	0.3125	1	0.0625	0	0
AA	A/G	7	0.4375	5	0.3125	3	0.2
AA	A/A	4	0.25	10	0.625	12	0.8
	Total	16		16		15	
C	G/G	28	0.1986	4	0.0282	0	0
C	A/G	80	0.5674	38	0.2676	14	0.0993
C	A/A	33	0.2340	100	0.7042	127	0.9007
	Total	141		142		141	
H	G/G	7	0.292	0	0	0	0
H	A/G	12	0.5	6	0.26087	9	0.375
H	A/A	5	0.208	17	0.73913	15	0.625
	Total	24		23		24	

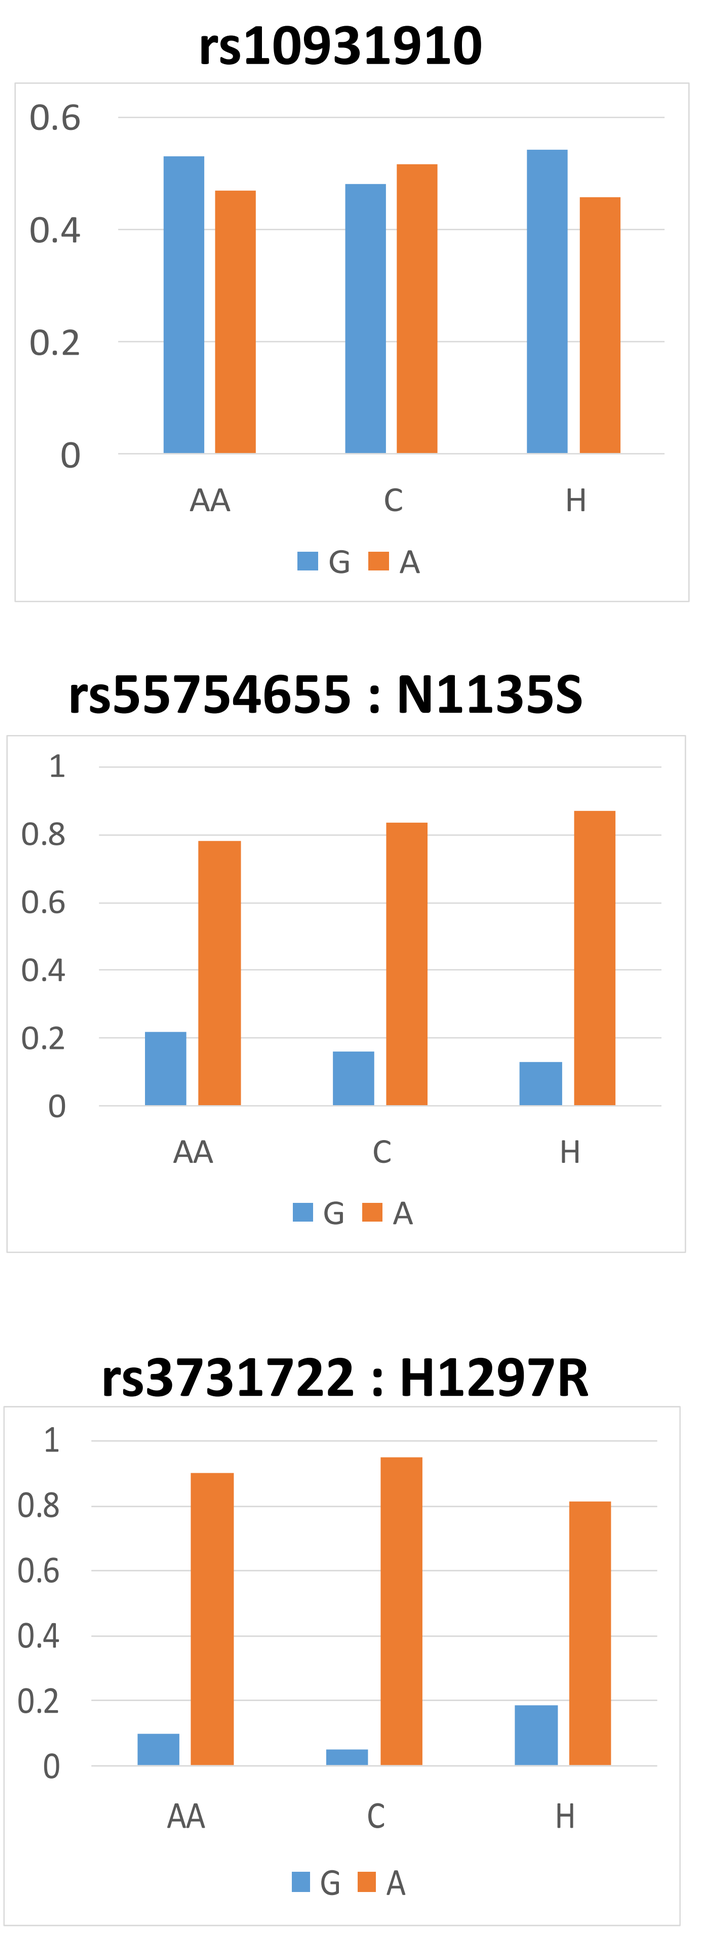
rs10931910
rs55754655 : N1135S
rs3731722 : H1297R
AA
C
H
G
A

Genotype data generated by Taqman PCR SNP assays were stratified by race and genotype frequencies calculated as a fraction of total. No significant differences were seen as tested by chi square analysis. Allelic frequencies were generated for each and graphed by race. Testing for rs3731722 revealed no homozygous mutants in any population. Low representation of both African American and Hispanic populations in the cohort may have limited the power of the analysis.

Figure 3.

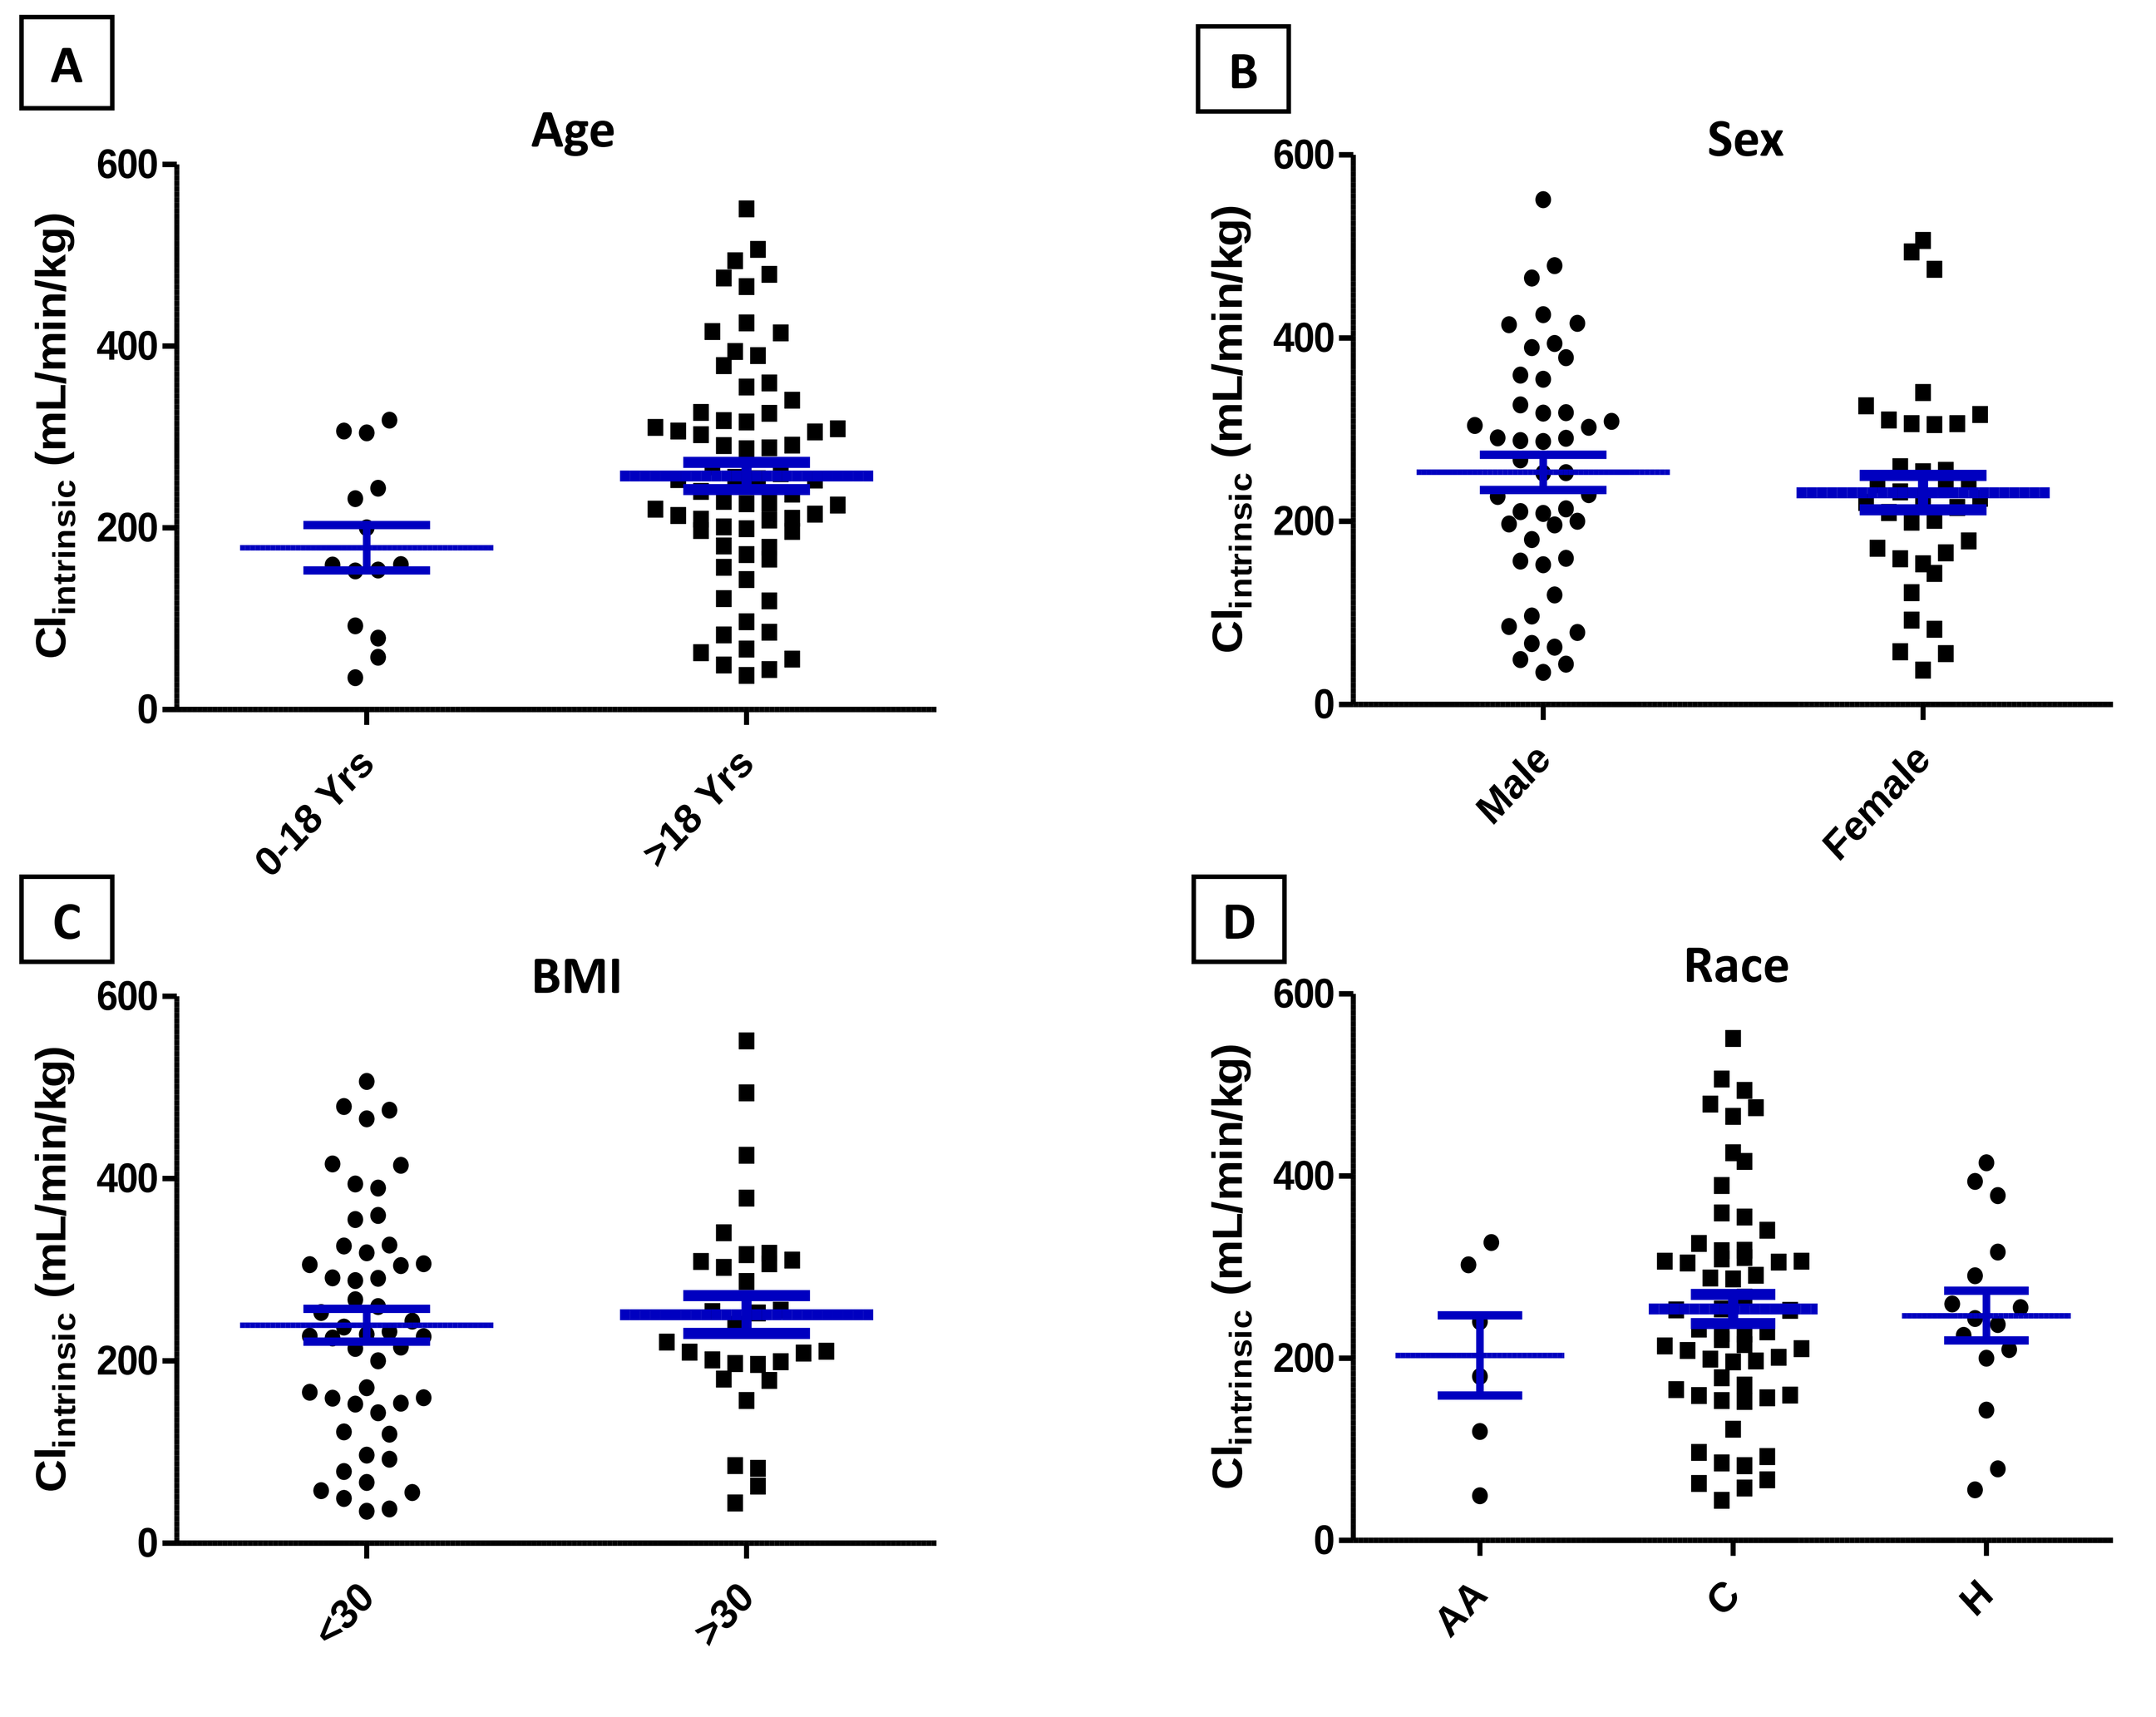
A
Age
0-18 Yrs
>18 Yrs
B
Sex
Male
Female
C
BMI
<30
>30
D
Race
AA
C
H
Clintrinsic (mL/min/kg)

Carbazeran *in vitro* hepatic clearance was stratified using donor demographics and whisker plots generated with mean+/-SEM. Statistical significance between groups was tested by t-test. Panel A shows values grouped by age in years , 0-18Yrs group was statistically different from the >18Yr group with mean activities, 178.2 vs 257.4 ml/min/kg, p=.0254 (n=14 and 67 respectively). Age differences have been reported in the literature with reduced expression prior to 1yro. (Tayame, 2007) Stratification by Sex (male n=45, female n=36) did not show a statistical difference, p=.4156. BMI did not statistically impact AO activity, using a BMI below and greater than 30, to separate non-obese and obese individuals ,mean clearance was 239.3 vs 250.9, p=.6795 (<30 n=50, >30 n=31). Clearance values were grouped by reported race data for African American (AA), (n=6), Caucasian (C) (n=58) and Hispanic (H) (n=15) and mean values genereated, 203.3, 254.2 and 246.9 ml/min/kg respectively. There was no statistical difference between the racial groups, AA vs C p=.3336, AA vs H p=.4026, C vs H p=.8341.

Figure 4.

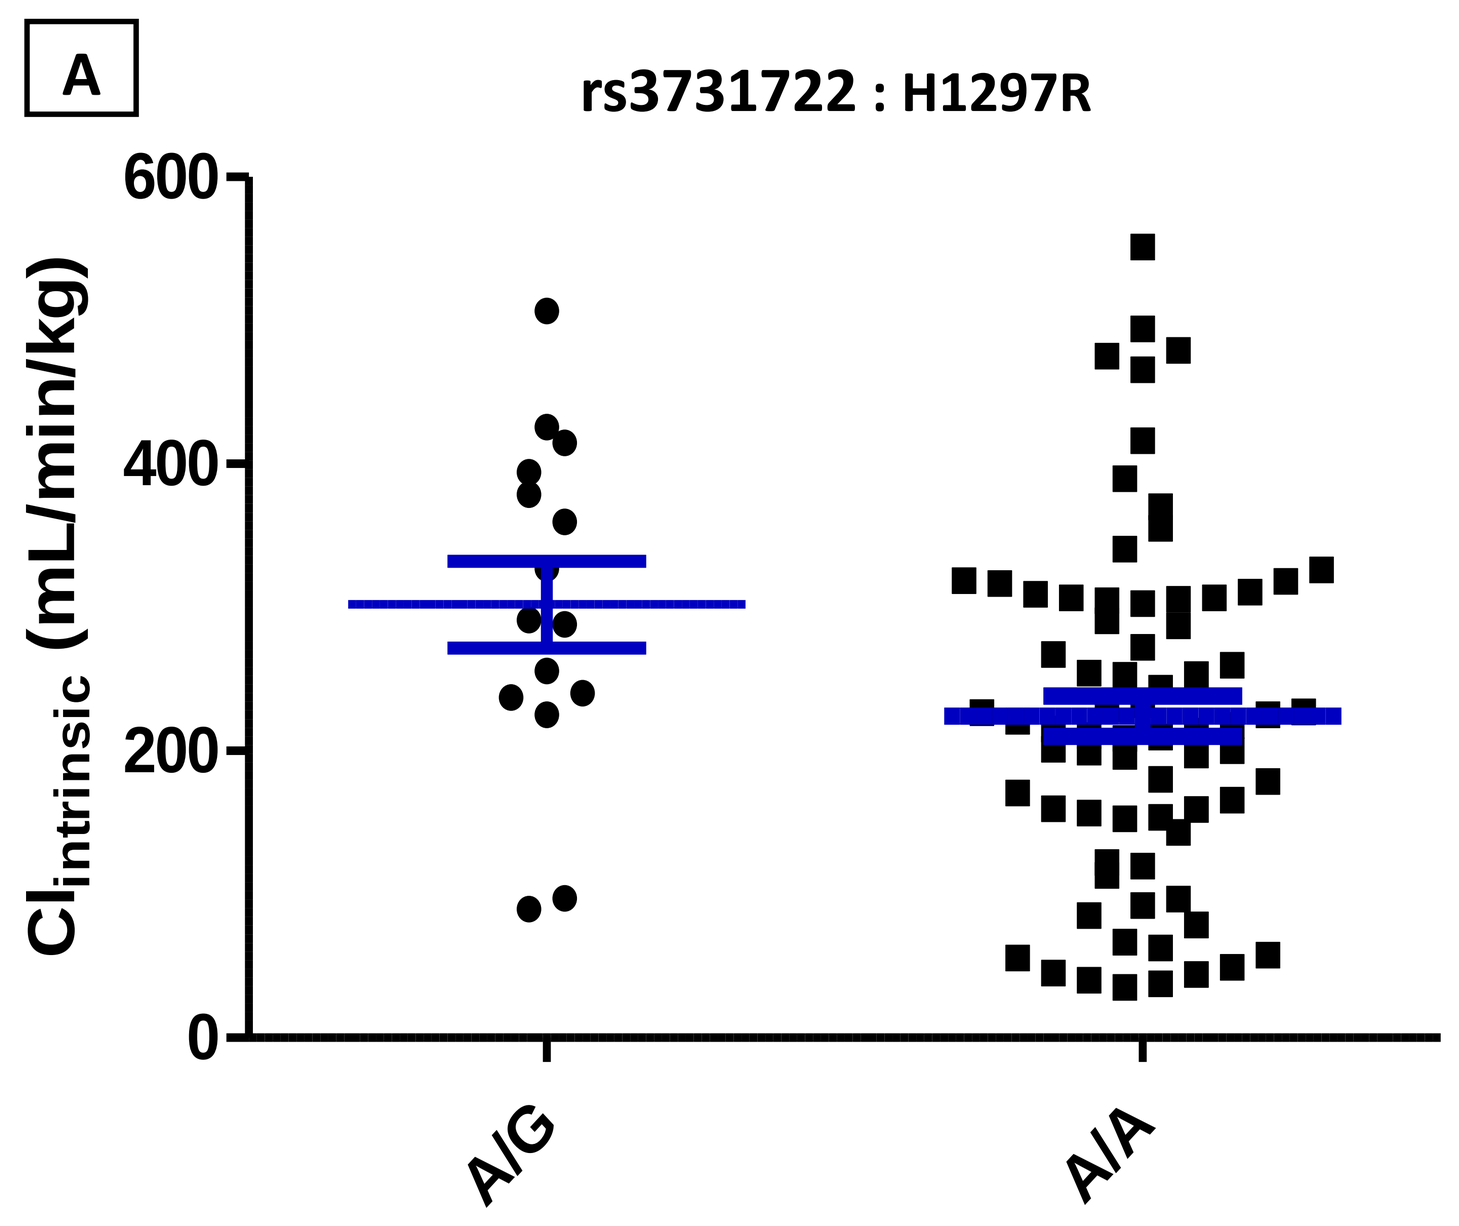
A
rs3731722 : H1297R
A/G
A/A
Clintrinsic (mL/min/kg)

A.

Genotpye data for SNP rs3731722 was generated and associated with the corresponding carbazeran clearance data for each lot. Homozygous mutant (G/G) samples were not found in this cohort. Means between the A/G genotype (302.18 ml/min/kg, n=15) and A/A wildtype genotype (224.29 ml/min/kg, n=74) were statistically different (p=.02326) between the two groups.

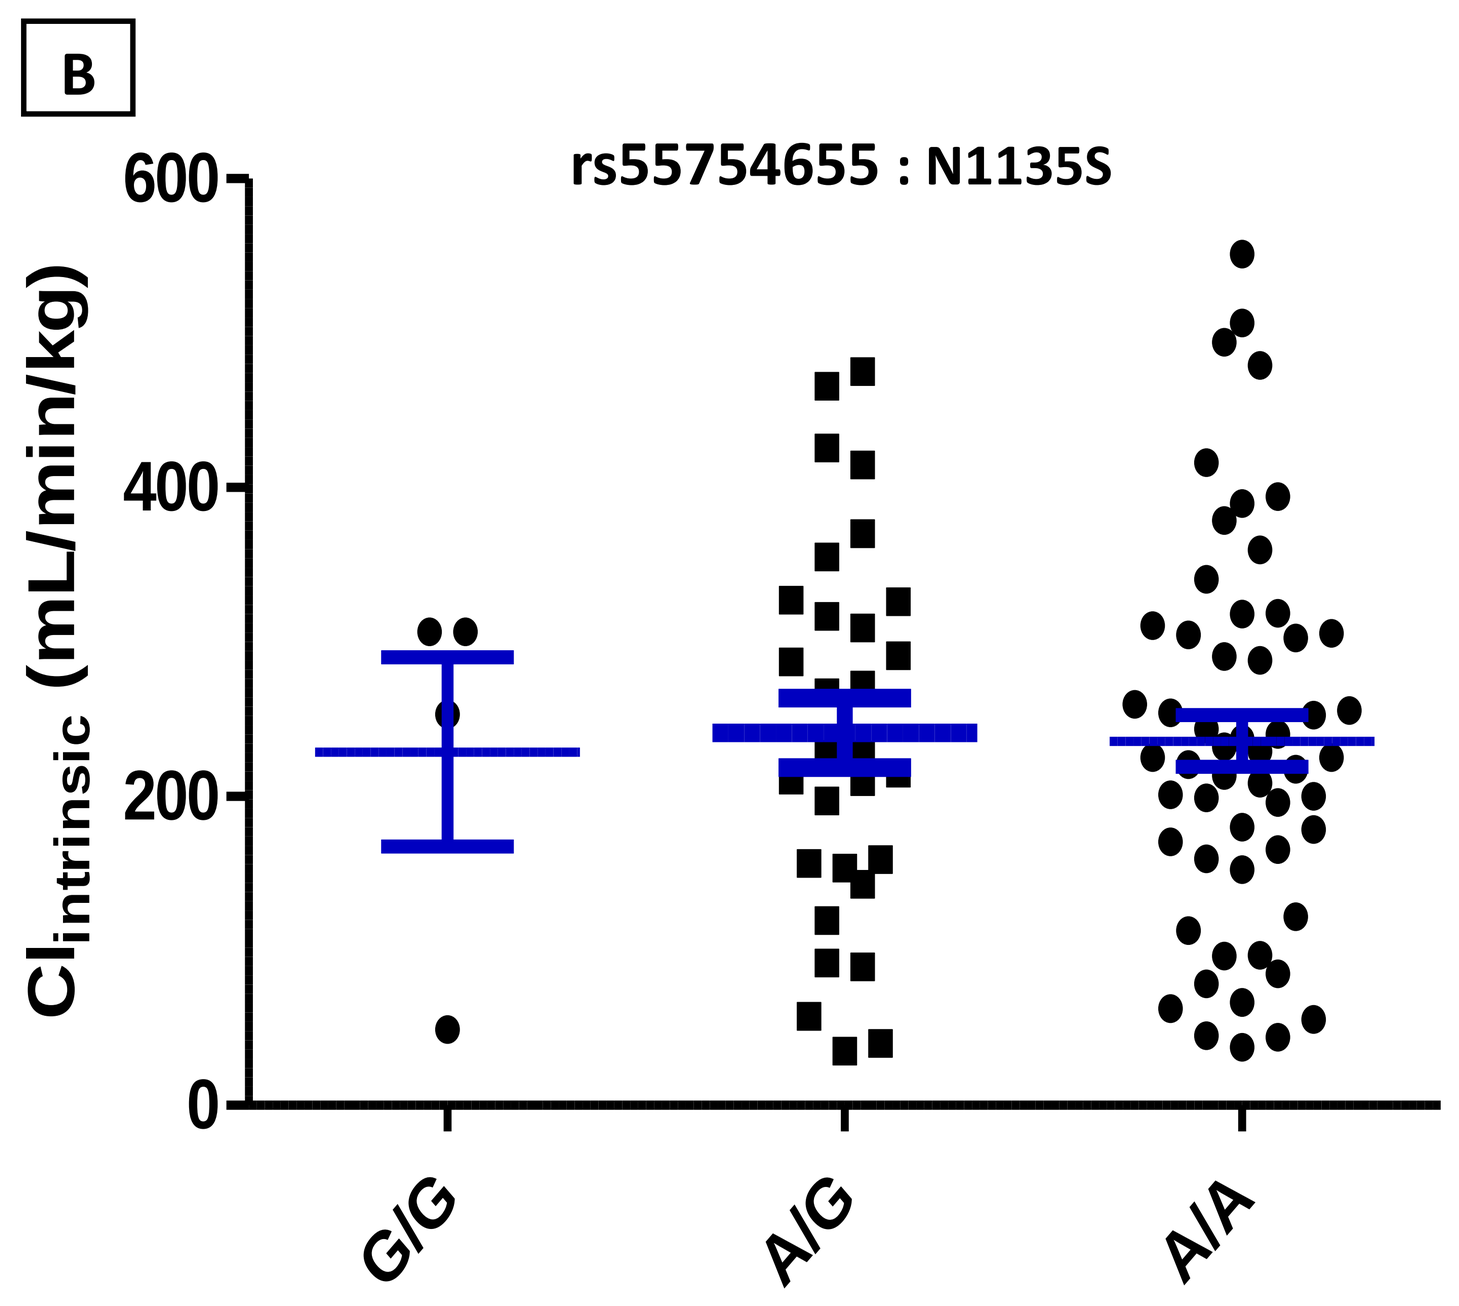
B
rs55754655 : N1135S
G/G
A/G
A/A
Clintrinsic (mL/min/kg)

B.

Genotpye data for SNP rs55754655 was generated and associated with the corresponding carbazeran clearance data for each lot. Homozygous mutant (G/G) samples infrequent in this cohort, but included for analysis. Means between the A/A genotype (228.91ml/min/kg, n=4), the A/G genotype (241.28 ml/min/kg, n=30), and G/G genotype (235.93 ml/min/kg, n=55) were not different between the three groups. A t-test was run for G/G vs A/A (p=.9128) and A/G vs A/A (p.8491) and neither was significant.

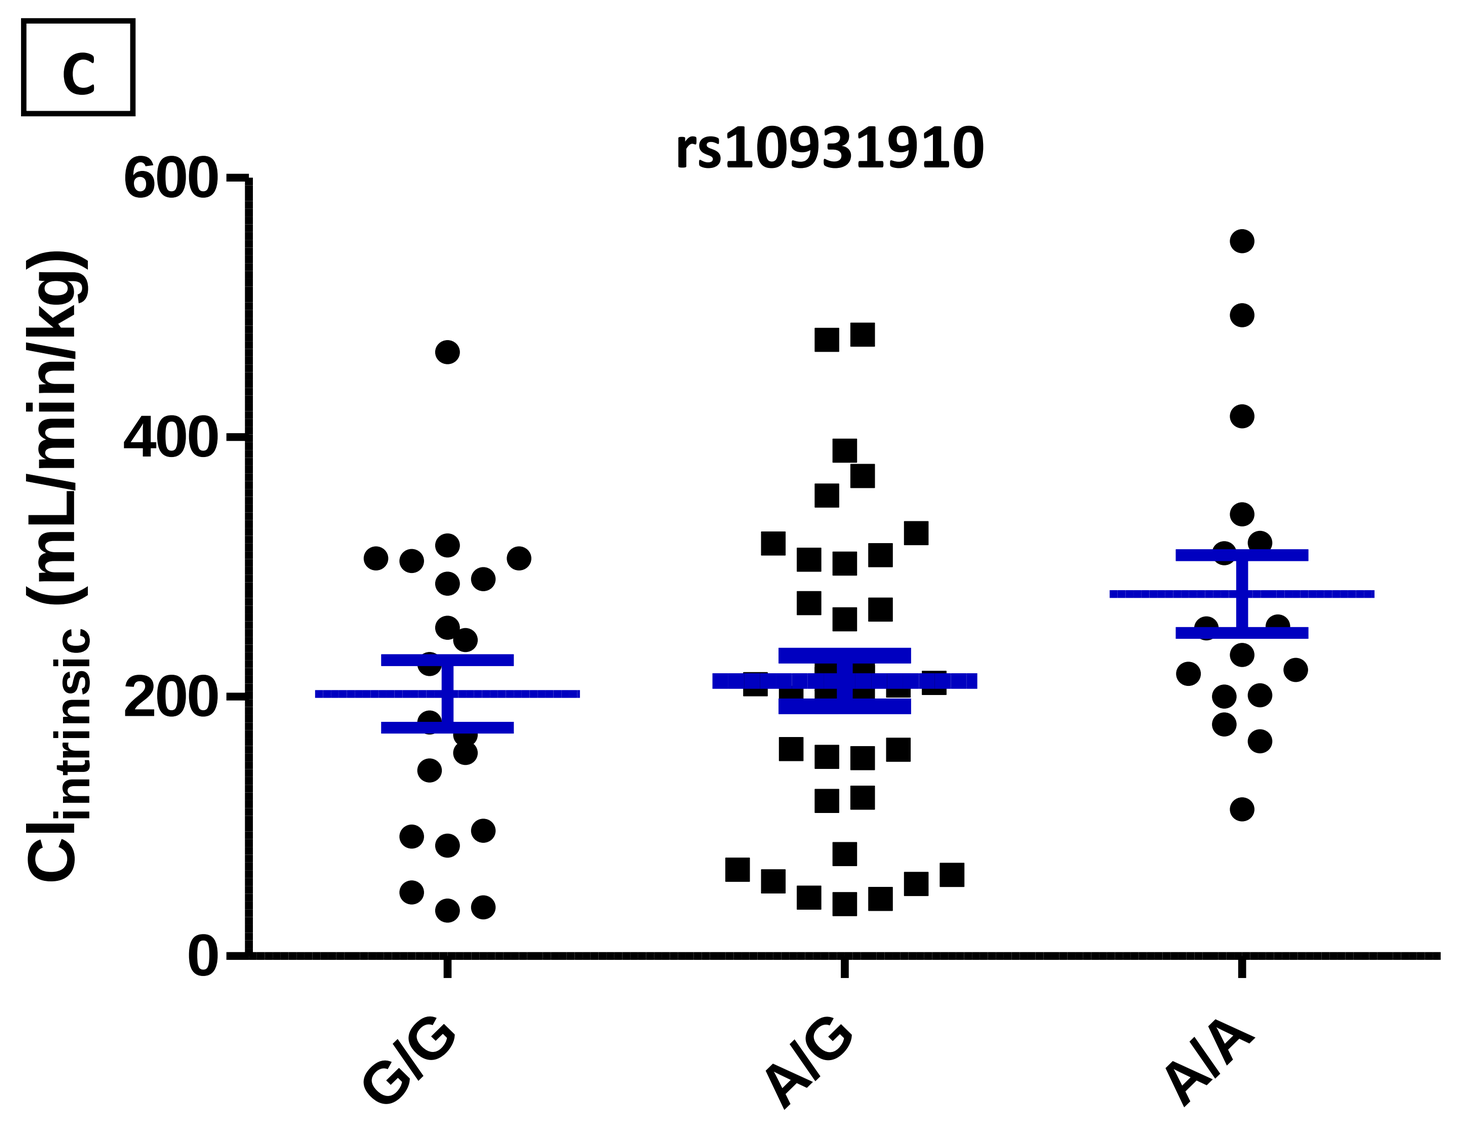
C
rs10931910
G/G
A/G
A/A
Clintrinsic (mL/min/kg)

C.

Genotpye data for SNP rs10931910 was generated and associated with the corresponding carbazeran clearance data for each lot. For the analysis haplotypes which contained rs3731722 G alleles were excluded from the analysis to isolate the impact of rs10931910. Means between the A/G genotype (212.26 ml/min/kg, n=37) and A/A genotype (279.23 ml/min/kg, n=16) were statistically different (p=.03789) between the two groups. However the G/G genotype mean (202.34 ml/min/kg, n=20) was not significantly different from the A/A genotype samples (p=.06107) possibly due to the small sample number and high variability.

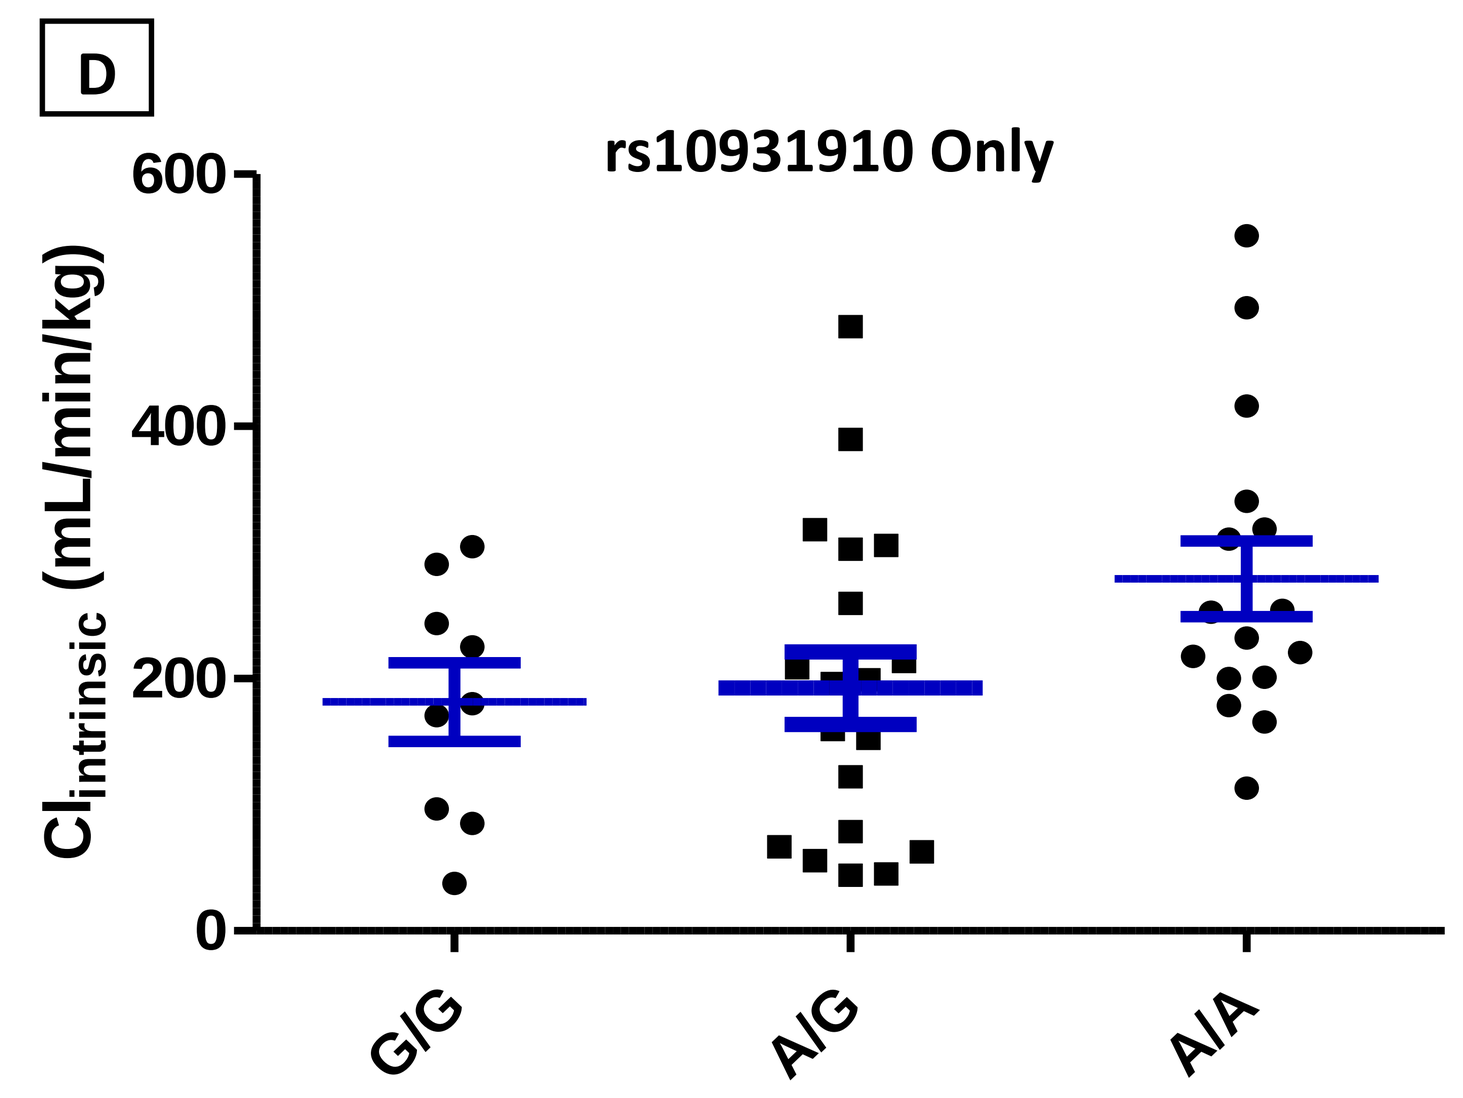
D
rs10931910 Only
G/G
A/G
A/A
Clintrinsic (mL/min/kg)

D.

The genotypes were further stratified by haplotype removing all rs55756455 "G" or mutant alleles from the cohort. Homozygous mutant (G/G) samples were not found in this cohort. Means between the A/G genotype (192.70 ml/min/kg, n=19) and A/A genotype (279.23 ml/min/kg, n=16) were statistically different (p=.04659) between the two groups. The G/G genotype mean (181.6 ml/min/kg, n=9) was significantly different from the A/A genotype samples (p=.04792). Though samples sizes were reduced. This haplotype analysis may shed light on the difficulty in determining SNP effects in clinical cohorts where "compensatory" SNPs may mask the effect of a single SNP .

Conclusions

- The single nucleotide polymorphisms rs10931910, rs55754655, and rs3731722 are present in human livers at high to low frequency respectively. The polymorphisms rs199984835, rs41309768, rs56199635, rs141786030 were monomorphic in our population.
- Racial differences were not statistically significant though small differences were detected.
- Donors under the age of 18 showed lower carbbazeran clearance than those over 18. Race, BMI and sex did not impact carbazeran clearance in hepatocytes.
- Rs3731722 showed statistically significant elevation in carbazeran clearance in hepatocytes heterozygous for the allele.
- Rs55754655 did not show a statistically significant difference in carbazeran clearance in our samples.
- Rs10931910 showed a statistically significant effect of the mutant allele in haplotypes which excluded rs3731722 and rs55754655.

Future Directions:

- Future studies would benefit from additional donors to improve statistical relevance of low frequency SNPs.
- Activity across a broader array of aldehyde oxidase substrates could provide insight into structure function relationships.
- A deeper haplotype analysis could identify the impact of individual SNPs on aldehyde oxidase function.